Inhaled cisplatin induces dose-dependent anti-tumor activity *in vivo* in an orthotopic lung cancer model.

<u>Lionel Larbanoix</u>[1], Tamara Davenne[2], Rémi Rosière [2], Sophie Laurent[1].

[1] Center for Microscopy and Molecular Imaging (CMMI), UMONS, Rue Adrienne Bolland 8, Gosselies, Belgium.
[2] InhaTarget Therapeutics, Rue Antoine de Saint-Exupery 2, Gosselies, Belgium

lionel.larbanoix@umons.ac.be

Introduction

Chemotherapy remains the backbone of treatment in most lung cancer patients at most of stages of the disease. Inhaled chemotherapy is a promising strategy to target lung tumors and limit the induced severe systemic toxicities observed with intravenous injection of cisplatin. Cisplatin dry powder for inhalation (CIS-DPI) was previously developed to allow sustained release and retention of cisplatin in the lungs and increases drug exposure at the targeted tumour site [*Chraibi S. et al., Eur J Pharm Biopharm. 2021 Jul; 164:93-104*].
This study investigates the anti-tumor activity of CIS-DPI activity in an orthotopic murine lung carcinoma model.

Methods

Under anesthesia, six weeks-old female BALB/c mice were grafted with 1.4 million M109-luc2 cells injected intercostaly.
In vivo bioluminescence imaging (BLI) of mice was performed twice a week after subcutaneous administration of 150 mg/kg of D-luciferin to follow tumor growth over time (a typical BLI is showed in Figure C).
On the 3rd day post-cell engraftment (i.e., day 3), mice with no detectable BLI signal were excluded, and the remaining mice were randomly allocated to different treatment groups.
The first treatment cycle started on day 7, which consisted of 5 days of CIS-DPI administration followed by 2 days of washout before starting the second and final CIS-DPI cycle (Figure A). Adapted CIS-DPI blends were manufactured and delivered to mice (under isoflurane anesthesia) using an endotracheal device (PADA, Aptar Pharma).

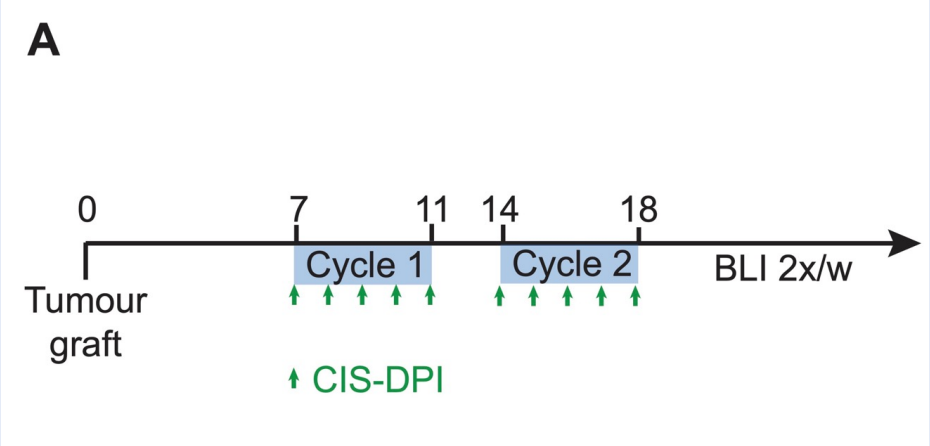

A
0
7
11
14
18
Cycle 1
Cycle 2
BLI 2x/w
Tumour graft
CIS-DPI

Figure A: Experiment schedule (days)

Results

A very significant reduction in tumor growth in all three CIS-DPI treated groups (0.1, 0.3 and 1 mg/kg cisplatin) was observed when compared with the vehicle and the untreated groups (Figure B). Moreover, a dose-dependent response was observed, with the strongest effect observed at the highest dose of 1 mg/kg cisplatin. Strikingly, the effect on tumors was rapid as the growth curves started separating before the end of the first treatment cycle, demonstrating the immediate antiproliferative action of cisplatin.

CIS-DPI significantly increased survival rate over time at 0.3 and 1 mg/kg compared with untreated and vehicle groups (Figure D). Specifically, the median survival was prolonged by +59%, from 18 days in the untreated and vehicle groups to 30.5 days in the CIS-DPI 1 mg/kg group.
In that same group, there were up to 40% responders on day 20, while this percentage was lower at the two lower dose levels (Figure E).

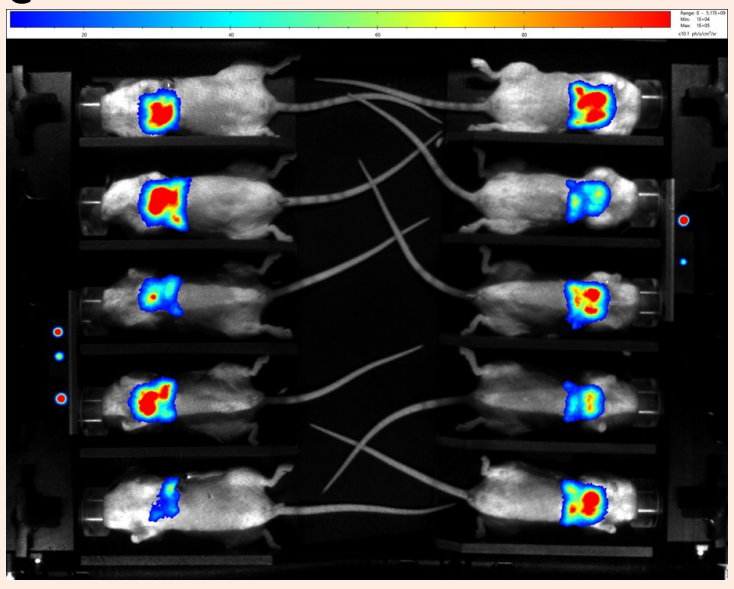

C

Figure C: Illustrative image of a typical BLI of 10 mice bearing lungs tumour.

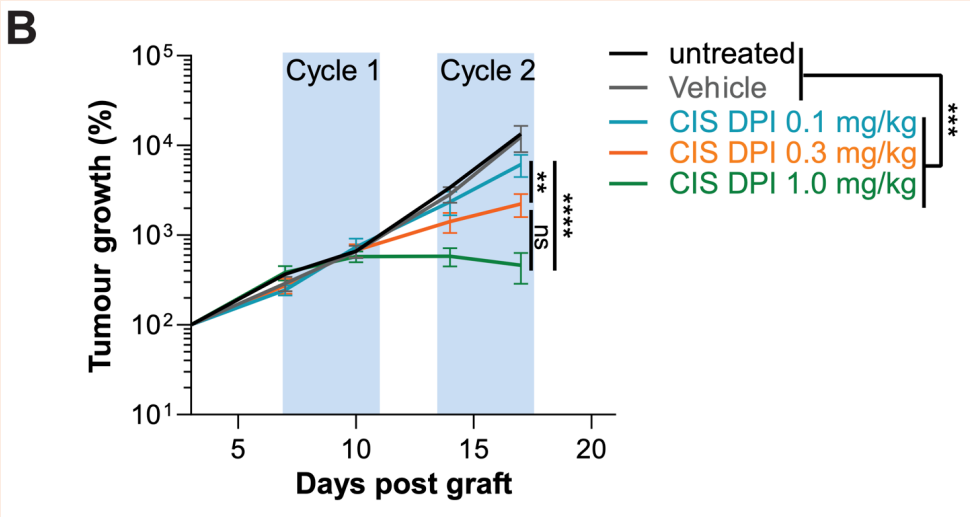

B
Cycle 1
Cycle 2
untreated
Vehicle
CIS DPI 0.1 mg/kg
CIS DPI 0.3 mg/kg
CIS DPI 1.0 mg/kg
Tumour growth (%)
Days post graft

**
ns

Figure B: Tumour growth was measured by BLI and expressed as % of signal measured at day 3, represented as a mean ± SEM. Untreated controls (*n* = 7), vehicle controls (*n* = 15), CIS-DPI 0.1 mg/kg (*n* = 18), CIS-DPI 0.3 mg/kg (*n* = 22) and CIS-DPI 1 mg/kg (*n* = 23). Statistic are two-way ANOVA multiple comparisons between groups at day 17.

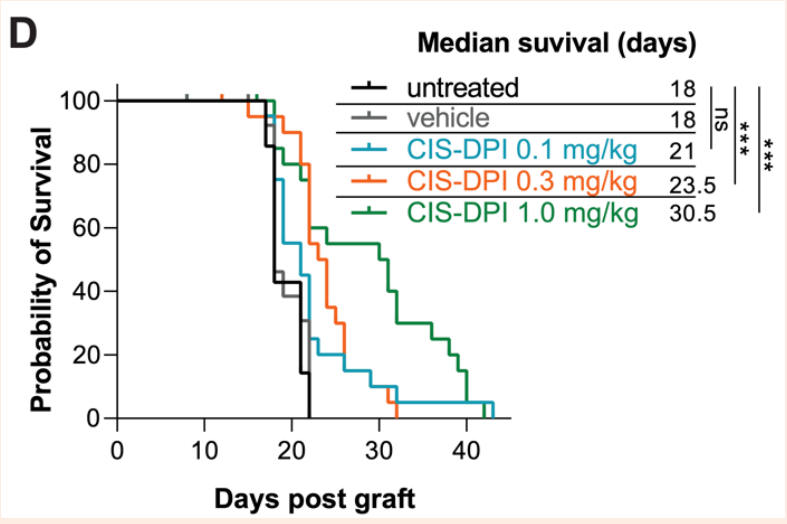

D
Median suvival (days)
untreated 18
vehicle 18
CIS-DPI 0.1 mg/kg 21
CIS-DPI 0.3 mg/kg 23.5
CIS-DPI 1.0 mg/kg 30.5
ns

Probability of Survival
Days post graft

Figure D: Kaplan-Meier survival curves and median survivals of mice (***$p < 0.001$)

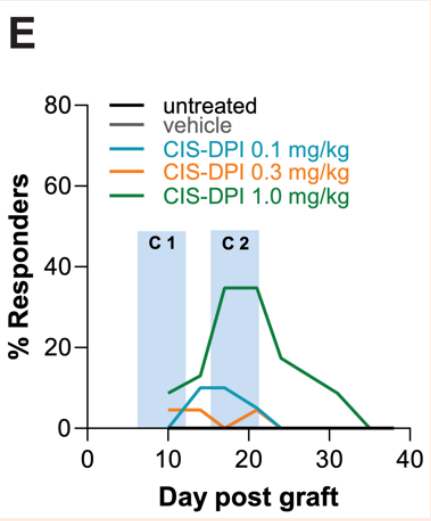

E
untreated
vehicle
CIS-DPI 0.1 mg/kg
CIS-DPI 0.3 mg/kg
CIS-DPI 1.0 mg/kg
C 1
C 2
% Responders
Day post graft

Figure E: At every time point, the proportion of responders in each group was calculated and plotted. An animal is considered responder if it has a BLI signal below 150% (100% being the signal at day 3).

Conclusions

Altogether, the results demonstrate a dose-dependent antitumor activity of CIS-DPI on M109 lung tumours, resulting in tumour shrinkage and prolonged median survival. Dosing at 1 mg/kg five times a week was the most potent CIS-DPI dose level. The use of a dry powder inhaler offers the possibility to deliver a smaller dose of chemotherapy every day compared with conventional systemic chemotherapy and opens the door to metronomic chemotherapy (MCT).
These results, and the efficacy study of CIS-DPI combination with the immune checkpoint inhibitor anti-PD1, were recently published [*Davenne T. et al., J Control Rel. 2023 Jan; 353:317-326*].

Link to the paper: